SRF231, a fully human CD47 antibody, potentiates the effects of opsonizing antibodies and cytotoxic chemotherapies in preclinical cancer models

Kshama A. Doshi, Matthew Rausch, Caroline M. Armet, Li Zhang, Alison M. Paterson, Benjamin H. Lee, Vito J. Palombella, Pamela M. Holland, Marisa O. Peluso

Surface Oncology, Inc., Cambridge, MA, USA

SURFACE ONCOLOGY™

Poster #2196

Background

- CD47, often referred to as a "Don't Eat Me" signal, is widely expressed on tumor cells and an important checkpoint of the innate immune response
- Several unique anti-CD47 molecules are currently being evaluated in the clinic to antagonize the CD47 axis in cancer
- SRF231 is a high-affinity, CD47-targeting antibody that blocks the SIRPα/CD47 interaction and induces FcR (CD32a)-dependent phagocytosis and cell death
- SRF231 also potentiates combinatorial activity of agents known to provide additional "Eat Me" signals to tumor cells such as:
 - Opsonizing hIgG1-containing antibodies that enhance Fc-FcR interactions
 - Chemotherapies that trigger cell death induction pathways

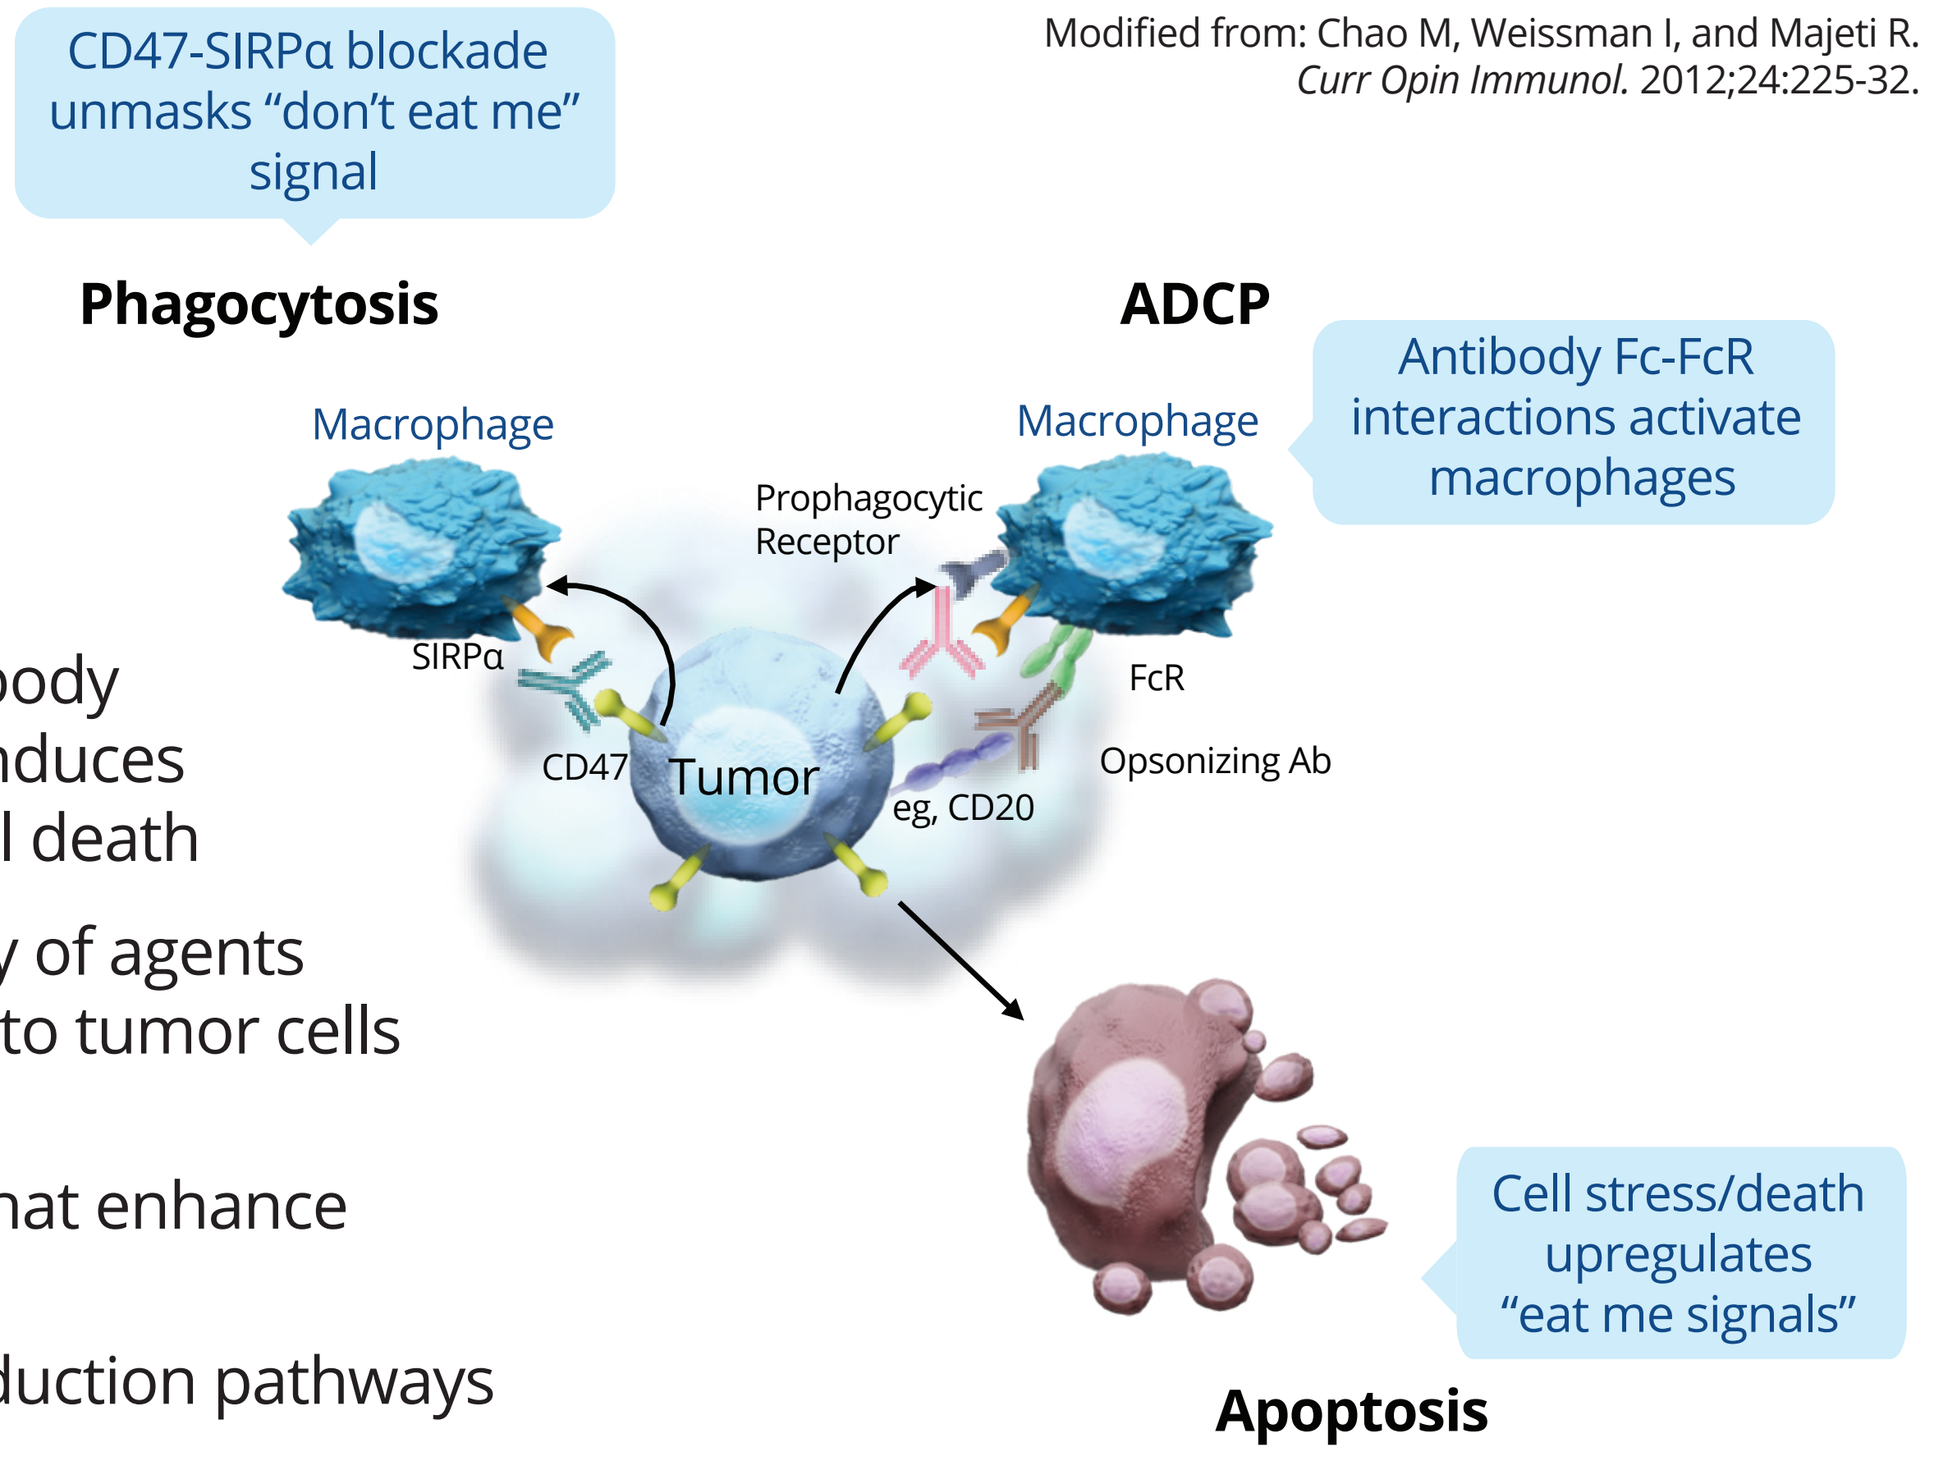
Modified from: Chao M, Weissman I, and Majeti R. *Curr Opin Immunol.* 2012;24:225-32.
CD47-SIRPα blockade unmasks "don't eat me" signal
Phagocytosis
ADCP
Antibody Fc-FcR interactions activate macrophages
Macrophage
Macrophage
Prophagocytic Receptor
SIRPα
FcR
CD47
Tumor
eg, CD20
Opsonizing Ab
Cell stress/death upregulates "eat me signals"
Apoptosis

SRF231 Potentiates the Effects of Opsonizing Antibodies and Chemotherapy In Vitro

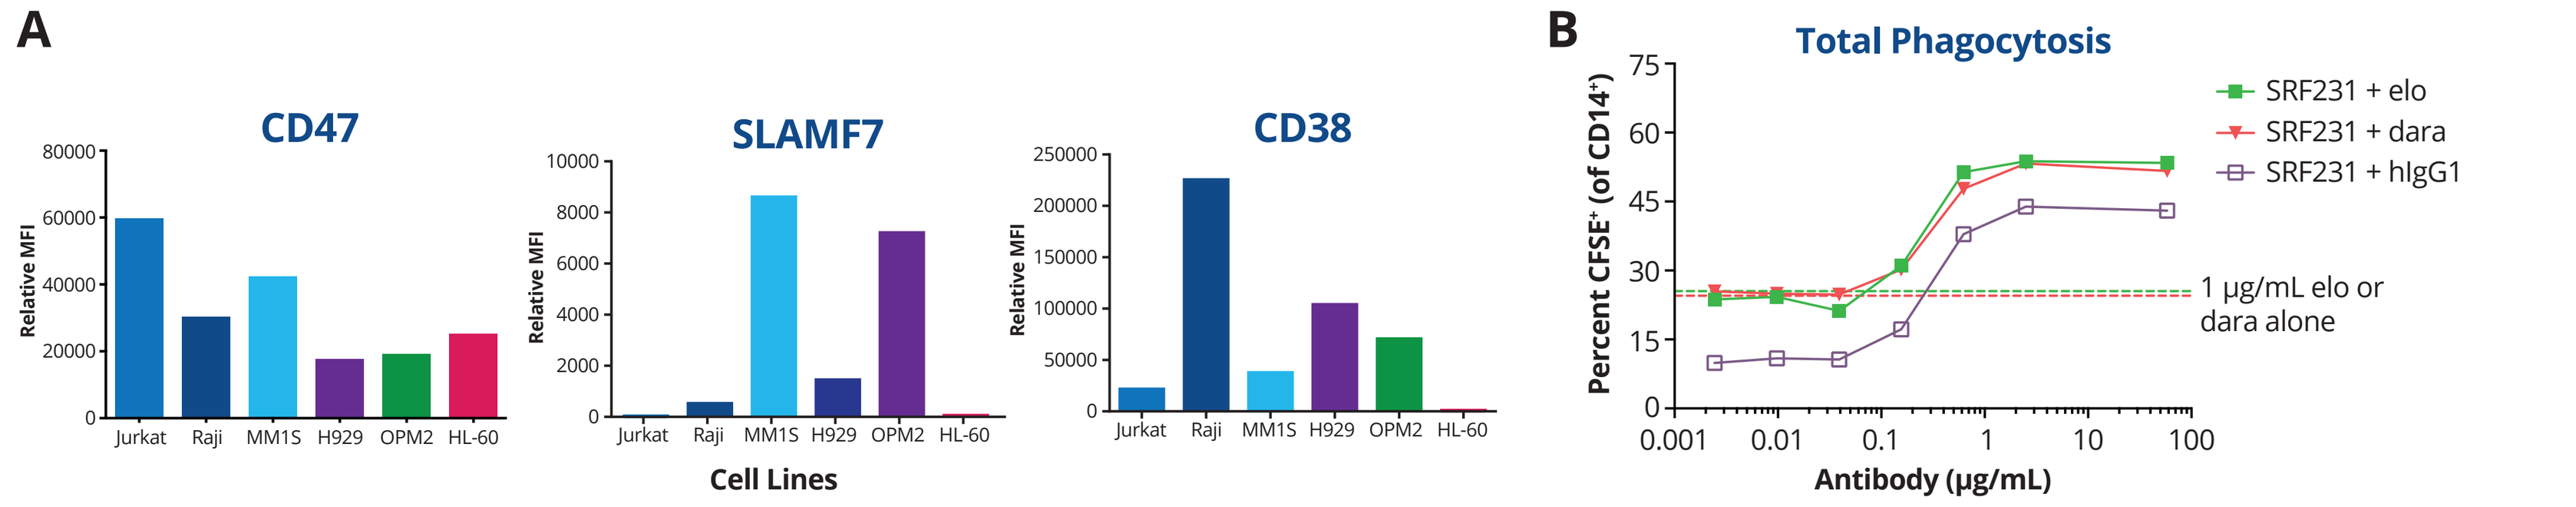
A
CD47
SLAMF7
CD38
Relative MFI
Jurkat
Raji
MM1S
H929
OPM2
HL-60
Cell Lines
B
Total Phagocytosis
SRF231 + elo
SRF231 + dara
SRF231 + hIgG1
1 µg/mL elo or dara alone
Percent CFSE⁺ (of CD14⁺)
Antibody (µg/mL)

Figure 1: SRF231 Potentiates Phagocytosis Induced by Opsonizing Agents, Elotuzumab (anti-SLAMF7), or Daratumumab (anti-CD38) in Multiple Myeloma

(A) Hematologic cancer cell lines were profiled for CD47, SLAMF7, or CD38 expression via flow cytometry. Relative mean fluorescence intensity (MFI) values reported (antigen MFI- isotype control MFI). **(B)** CFSE-labeled MM.1S multiple myeloma tumor cell targets were cultured with human monocyte-derived macrophages at a 2:1 target:effector ratio and incubated with a dose-response of SRF231 in the presence of a single dose of elotuzumab (elo) or daratumumab (dara, 1 µg/mL) or hIgG1 control (10 µg/mL) for 2 h at 37°C. Cells were stained with anti-CD14 and analyzed by flow cytometry. Phagocytosis denoted by percent CFSE⁺ tumor cells within CD14⁺ macrophages. Dotted lines indicate phagocytosis induced by 1 µg/mL elo or dara alone.

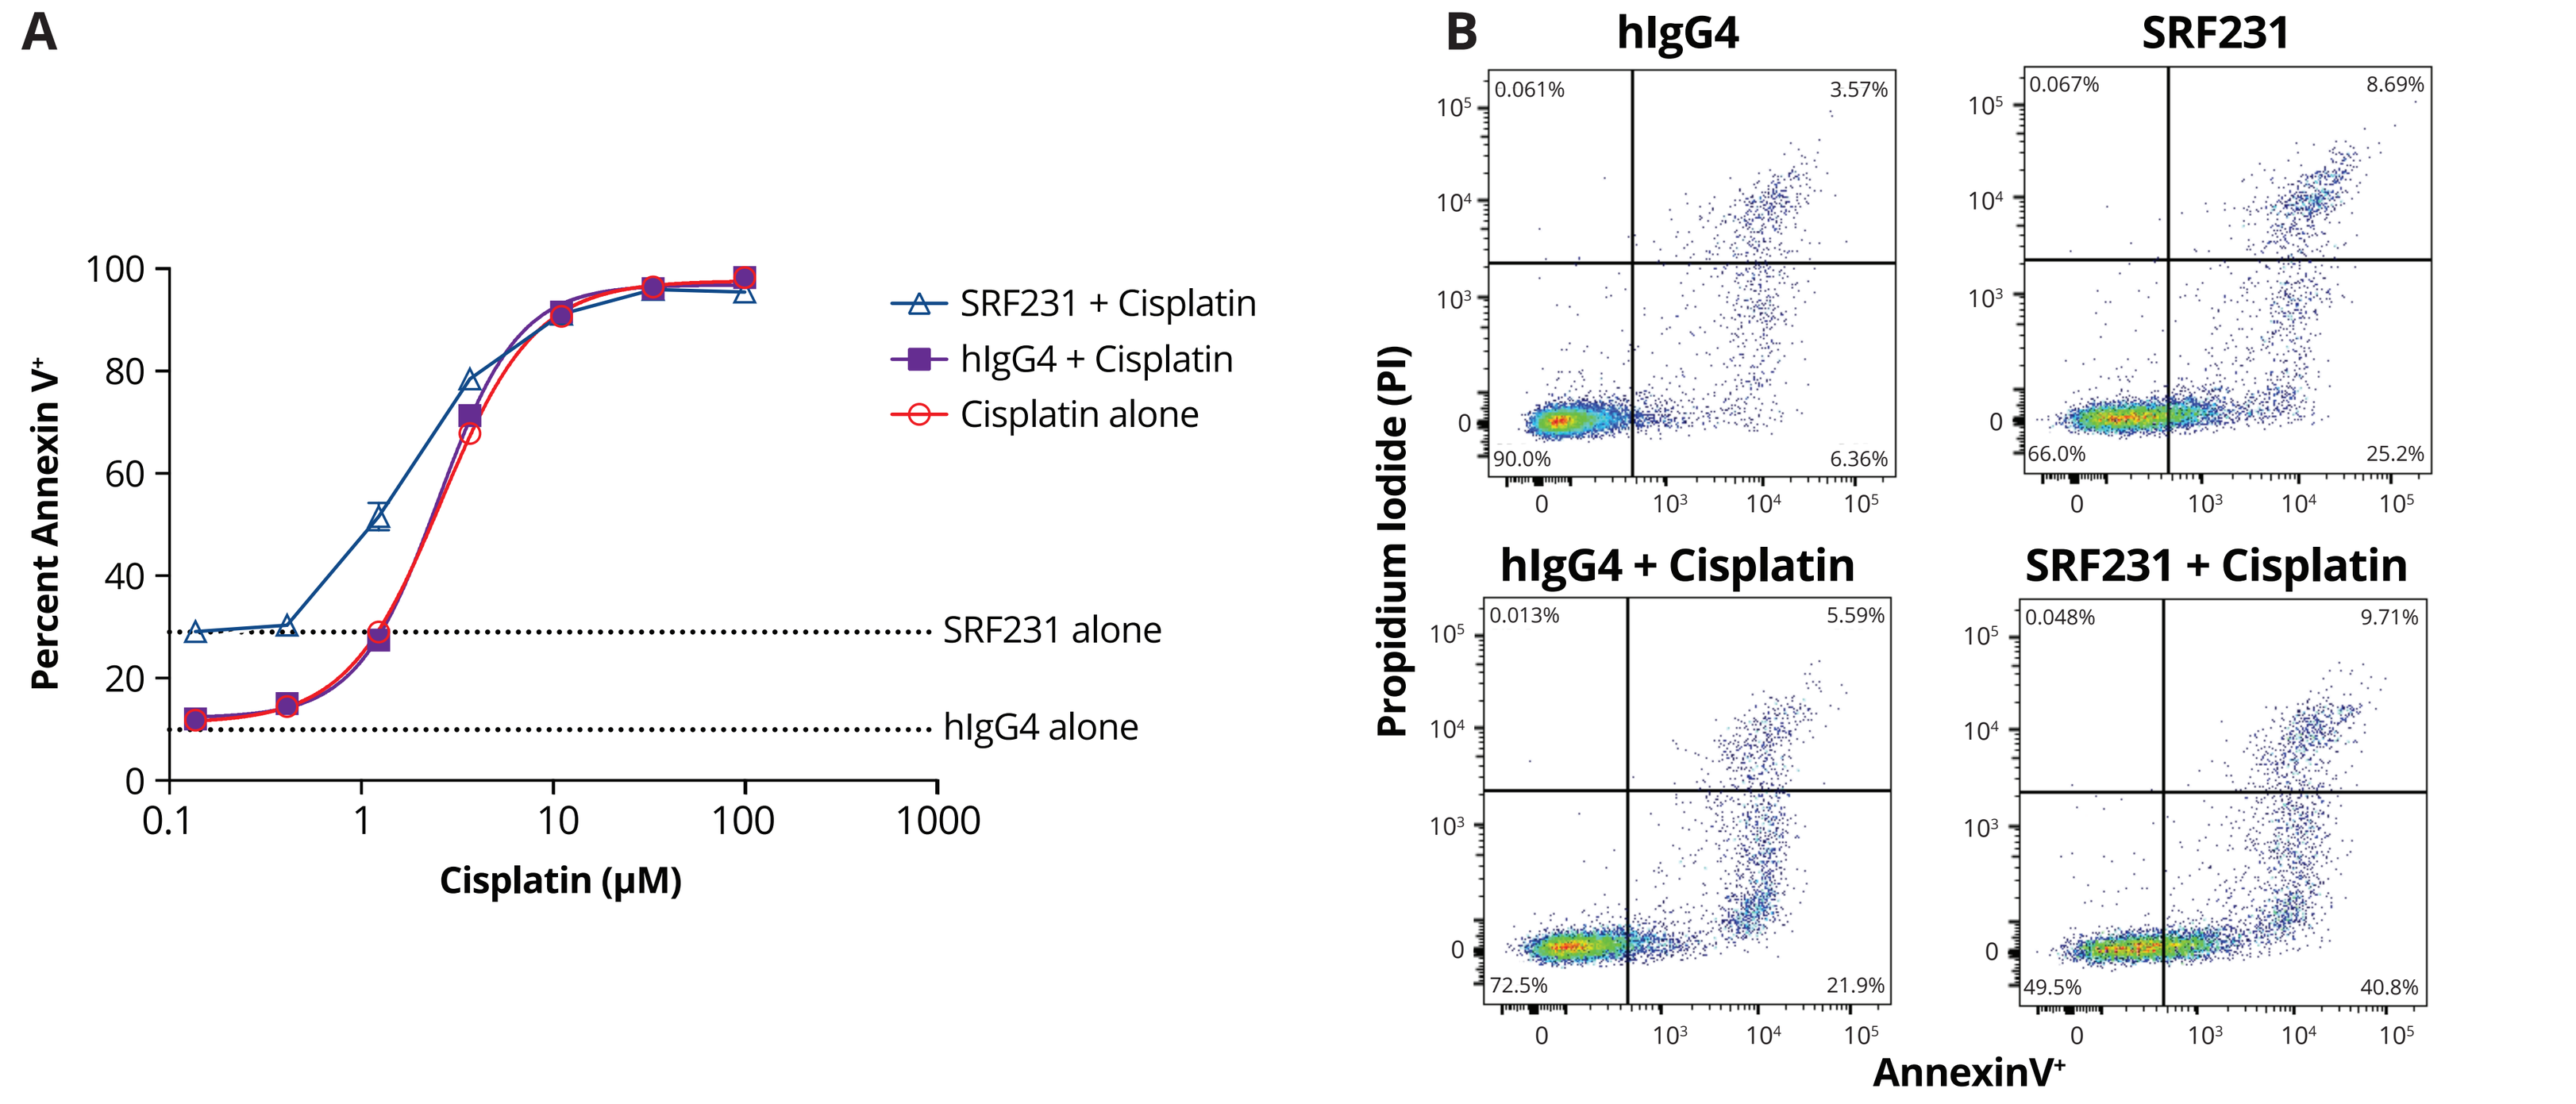
A
SRF231 + Cisplatin
hIgG4 + Cisplatin
Cisplatin alone
SRF231 alone
hIgG4 alone
Percent Annexin V⁺
Cisplatin (µM)
B
hIgG4
SRF231
hIgG4 + Cisplatin
SRF231 + Cisplatin
Propidium Iodide (PI)
AnnexinV⁺

Figure 2: SRF231 Potentiates Cell Death Induced by Cisplatin In Vitro

(A) Protein-G plates were precoated with 10 µg/mL SRF231 or hIgG4 antibody and incubated at 4°C overnight. Jurkat cells were then seeded on antibody-coated protein-G plates +/- a dose-response of cisplatin. Plates were incubated at 37°C for 24 h and Annexin V⁺ tumor cells assessed via flow cytometry. **(B)** Representative flow cytometry images corresponding to 10 µg/mL antibody alone (upper panels) or antibody + 0.4 µM cisplatin (lower panels).

SRF231 + Opsonizing Antibodies (Elotuzumab, Daratumumab) Leads to Enhanced Antitumor Activity in Multiple Myeloma Xenograft Models

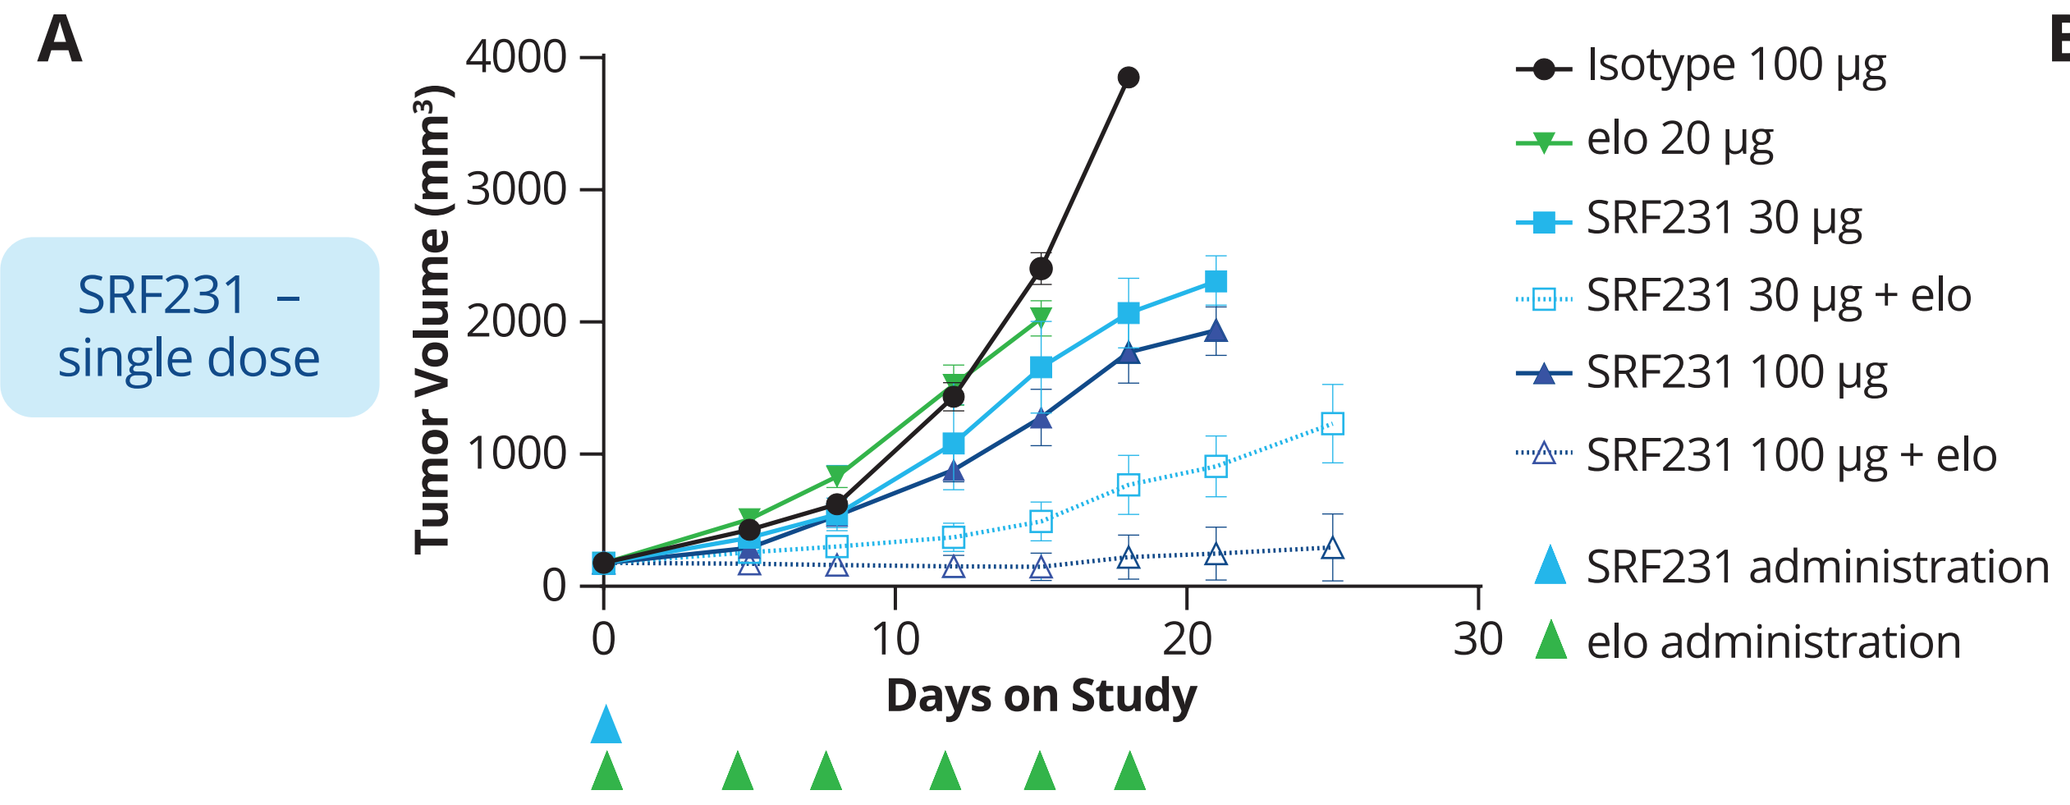
A
SRF231 – single dose
Tumor Volume (mm³)
Days on Study
Isotype 100 µg
elo 20 µg
SRF231 30 µg
SRF231 30 µg + elo
SRF231 100 µg
SRF231 100 µg + elo
SRF231 administration
elo administration

B

SRF231 Dose	CRR (D25) –elo	CRR (D25) +elo
0 µg	NA	0%
30 µg	0%	0%
100 µg	0%	44%

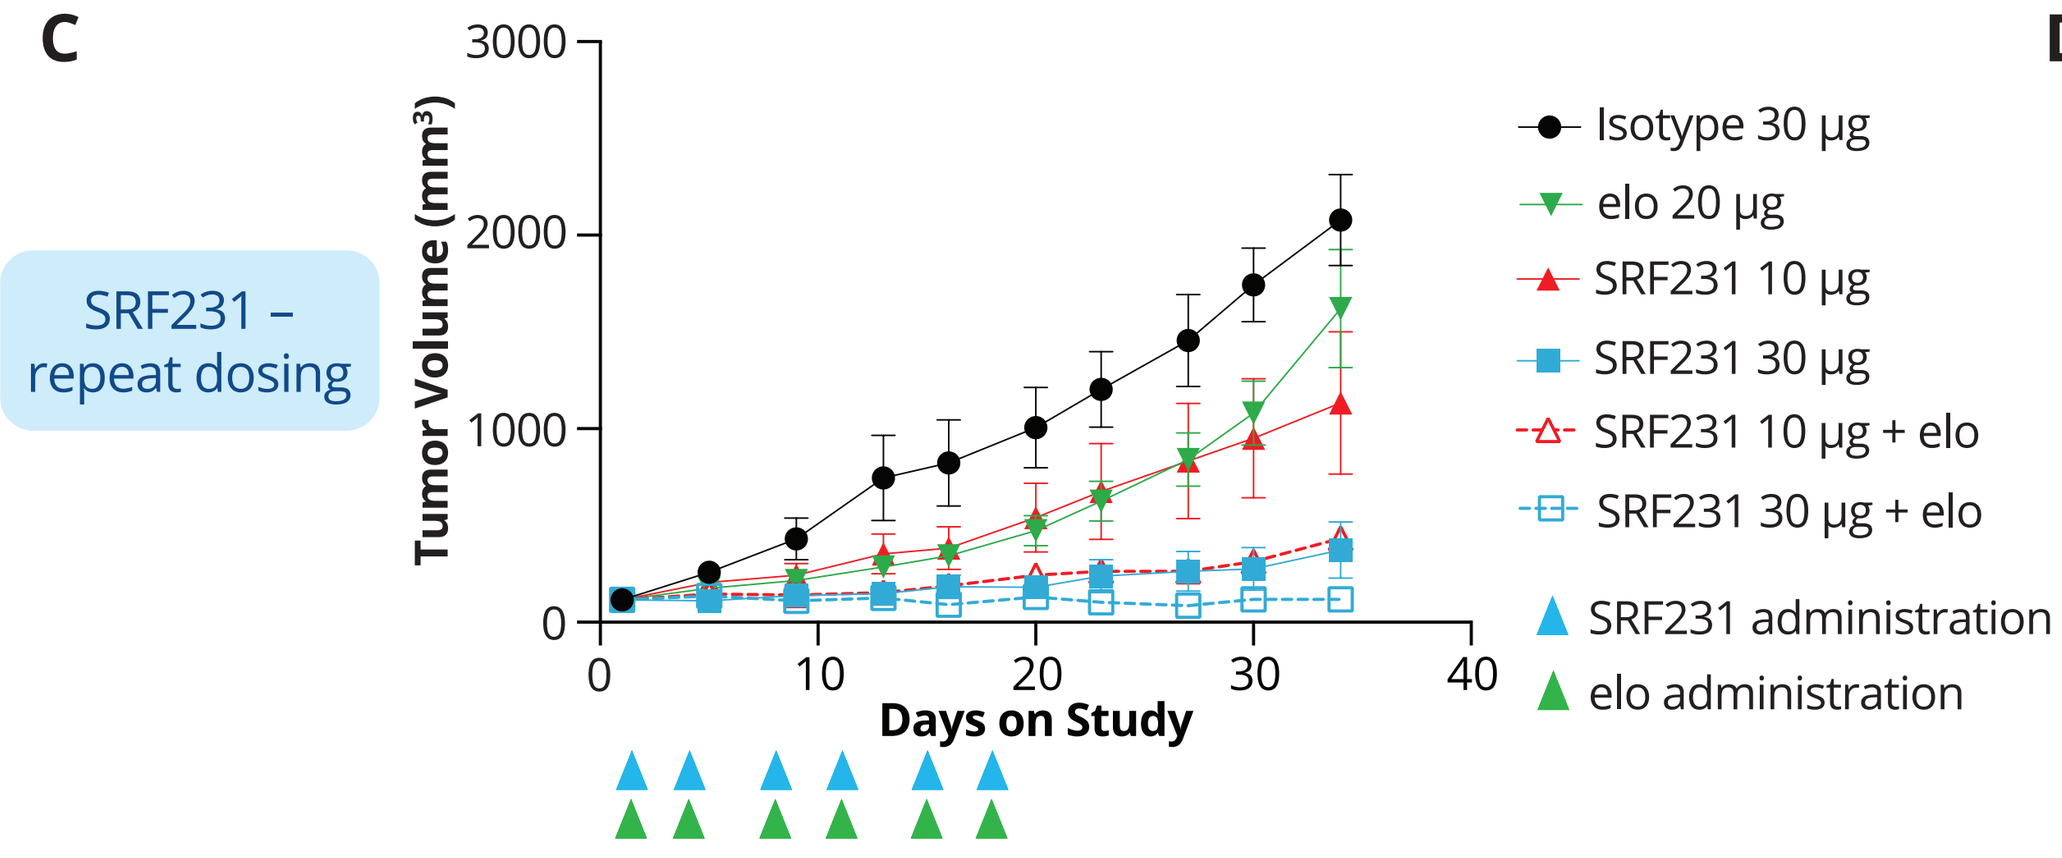
C
SRF231 – repeat dosing
Tumor Volume (mm³)
Days on Study
Isotype 30 µg
elo 20 µg
SRF231 10 µg
SRF231 30 µg
SRF231 10 µg + elo
SRF231 30 µg + elo
SRF231 administration
elo administration

D

SRF231 Dose	CRR (D34) –elo	CRR (D34) +elo
0 µg	NA	0%
10 µg	10%	30%
30 µg	30%	50%

Figure 3: Enhanced Antitumor Activity Observed in OPM2 Multiple Myeloma Xenograft Model with Elotuzumab + SRF231

OPM2 cells were inoculated subcutaneously (SC) into C.B-17 SCID mice. When tumors reached 100 mm³, mice were randomized and treated. **(A)** Animals were dosed intraperitoneally (IP) with 20 µg elo biw x 3 in addition to a single dose of SRF231 on Day 1. **(C)** C.B-17 SCID mice were implanted with tumors as described above and dosed IP with elo and SRF231 at the indicated doses and schedule. **(B,D)** Tables of complete response rate ([CCR], percentage of mice with no palpable tumors) observed with SRF231 +/- elo on Day 25 (single-dose study) or Day 34 (repeat-dose study), respectively.

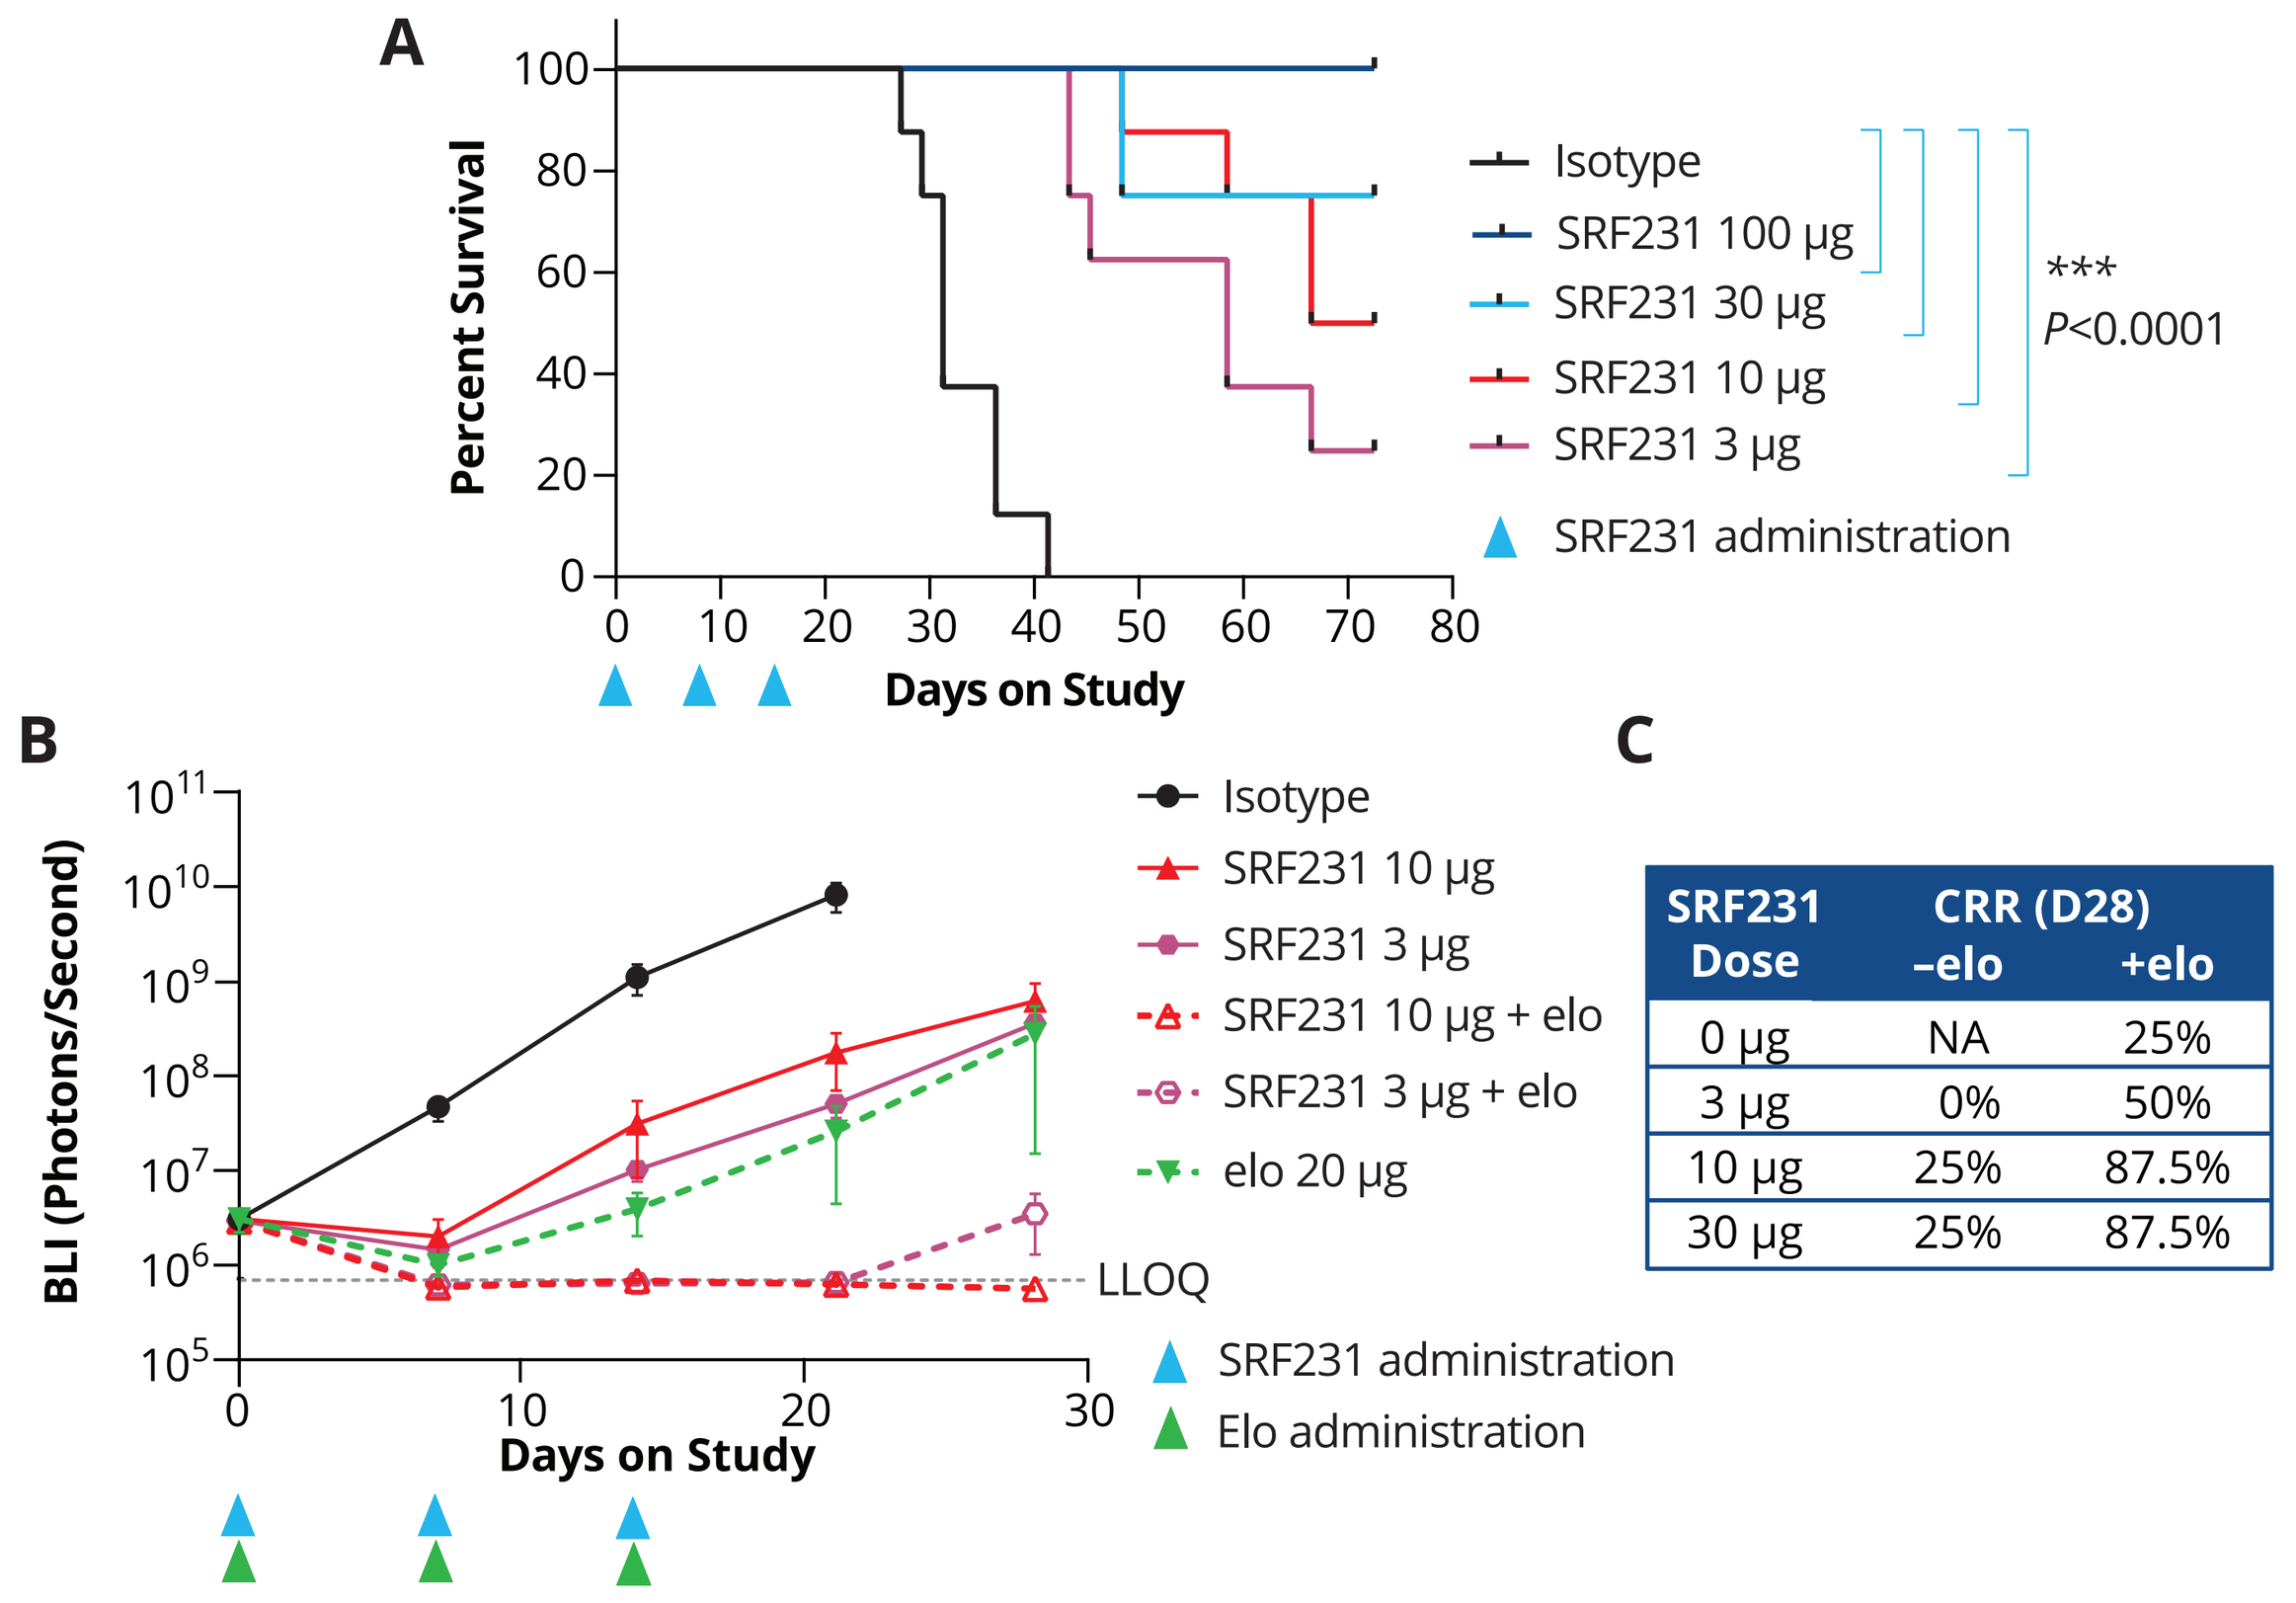
A
Percent Survival
Days on Study
Isotype
SRF231 100 µg
SRF231 30 µg
SRF231 10 µg
SRF231 3 µg
SRF231 administration

P<0.0001
B
BLI (Photons/Second)
Days on Study
Isotype
SRF231 10 µg
SRF231 3 µg
SRF231 10 µg + elo
SRF231 3 µg + elo
elo 20 µg
LLOQ
SRF231 administration
Elo administration

C

SRF231 Dose	CRR (D28) –elo	CRR (D28) +elo
0 µg	NA	25%
3 µg	0%	50%
10 µg	25%	87.5%
30 µg	25%	87.5%

Figure 4: Enhanced Antitumor Activity Observed with SRF231 +/- Elotuzumab in Disseminated MM.1S Multiple Myeloma Xenograft Model

C.B-17 SCID mice were injected IV with MM.1S-Luc tumor cells. Established disseminated disease was determined by whole-body bioluminescence imaging (BLI), and animals were treated with SRF231 +/- elo, IP, qw x 3. **(A)** Kaplan-Meier survival curve indicating SRF231 dose-dependent survival benefit (*P* values calculated by Log-rank [Mantel-Cox] test). **(B)** Efficacy plots indicating disease burden as measured by BLI (photons/second) in mice treated with SRF231 +/- elo. **(C)** Table of CRRs observed on Day 28.

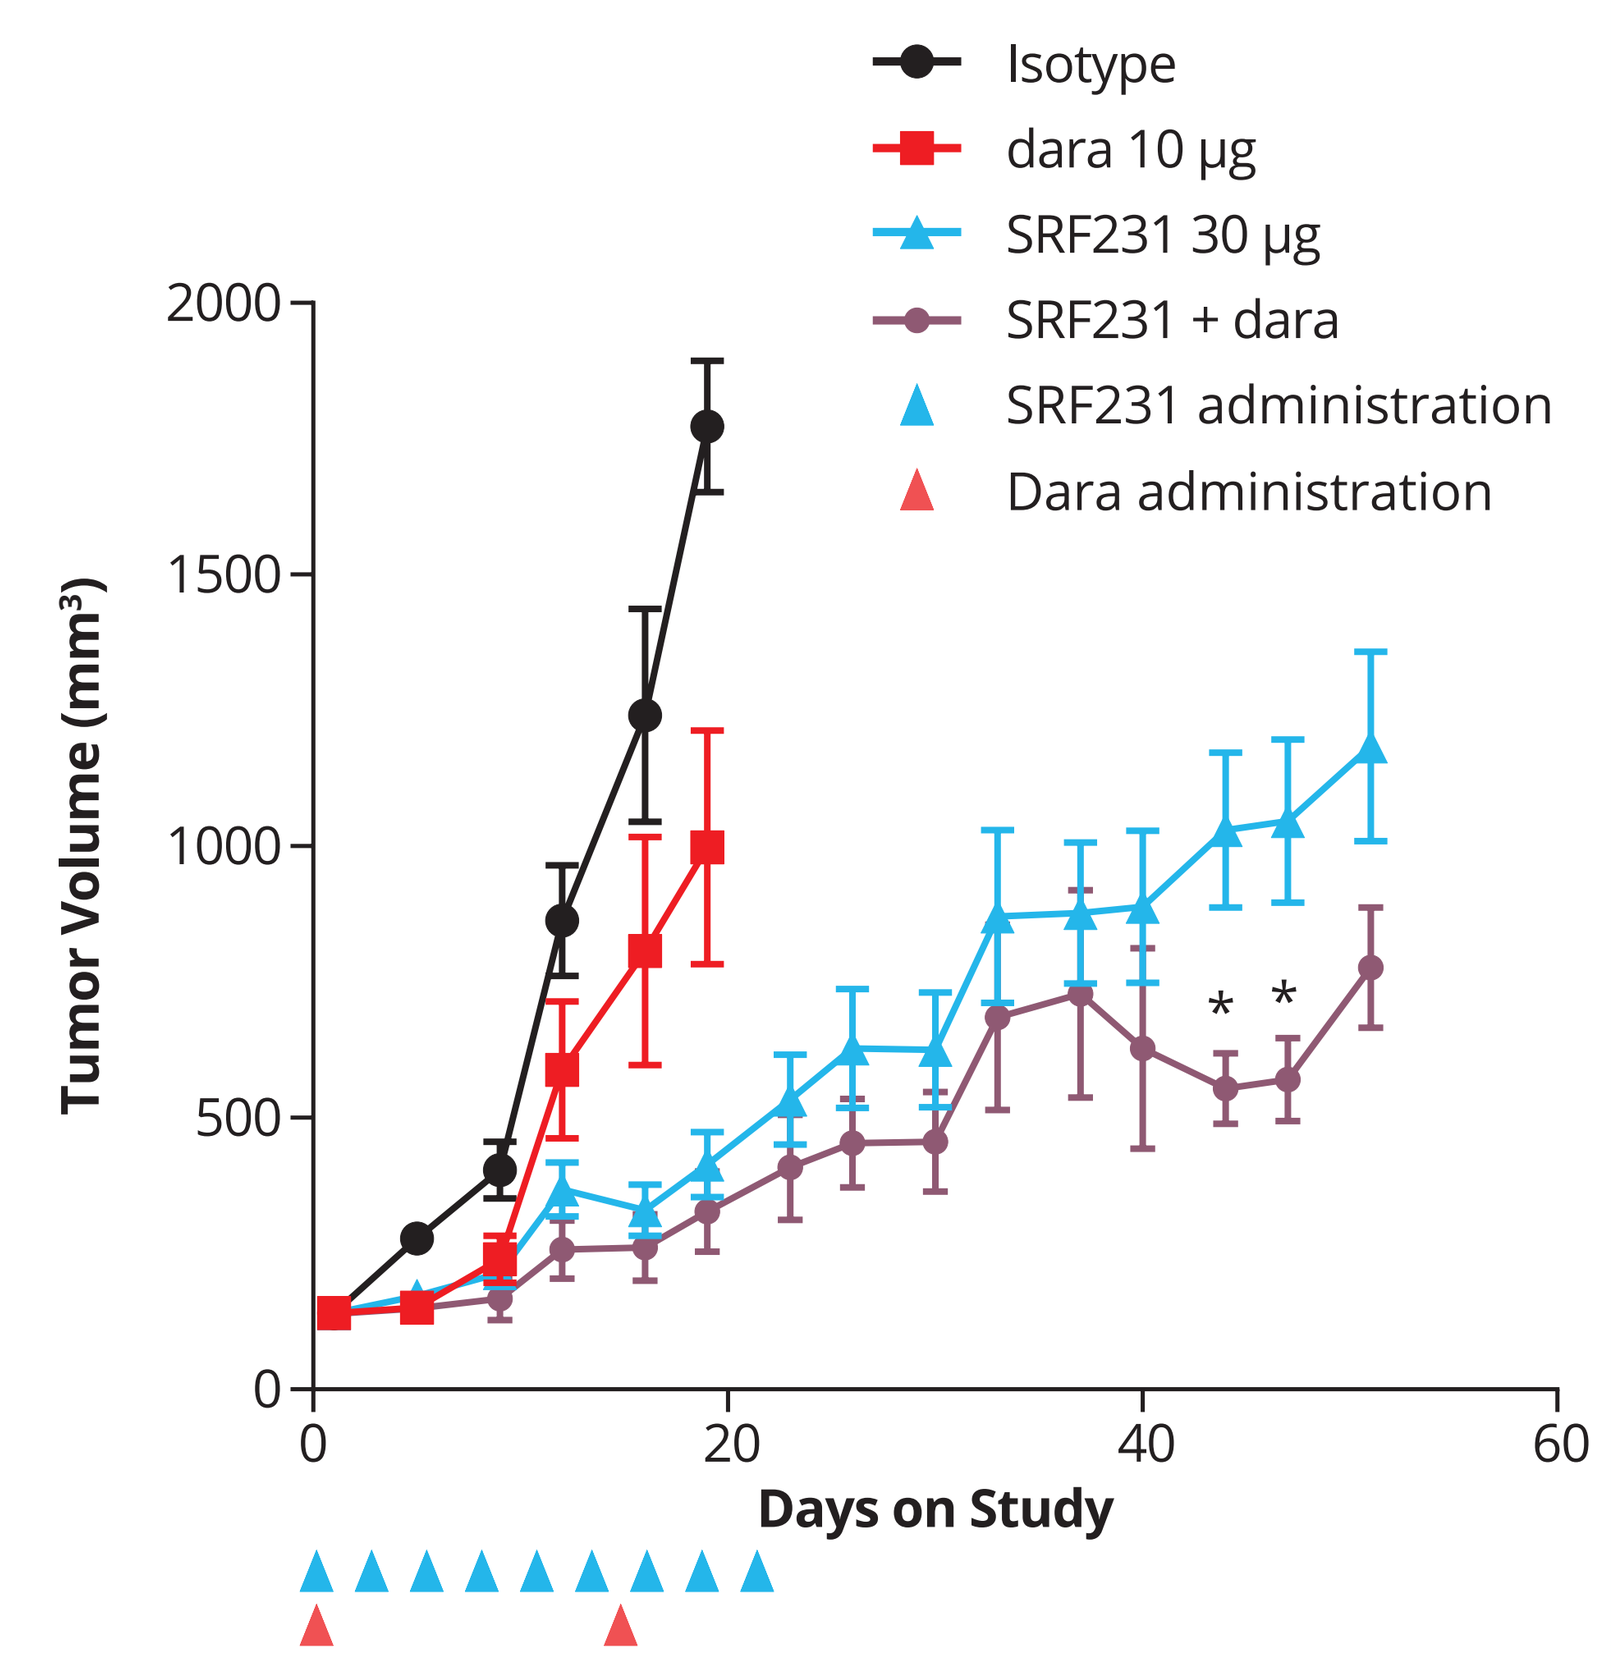
Isotype
dara 10 µg
SRF231 30 µg
SRF231 + dara
SRF231 administration
Dara administration
Tumor Volume (mm³)
Days on Study

Figure 5: Enhanced Antitumor Activity Observed with SRF231 +/- Daratumumab in H929 Xenograft Model

H929 tumor cells were implanted SC into CB.17 SCID mice. When tumors reached an average size of 150 mm³, mice were randomized and dosed IP with 30 µg SRF231 tiw x 3 and 10 µg dara on Days 1 and 15. Significant antitumor activity was observed in the combo arm relative to SRF231 alone on Day 44 and Day 47; **P*<0.05 (unpaired t-test).

SRF231 Potentiates the Effects of Chemotherapies in Lung Cancer Xenograft Models

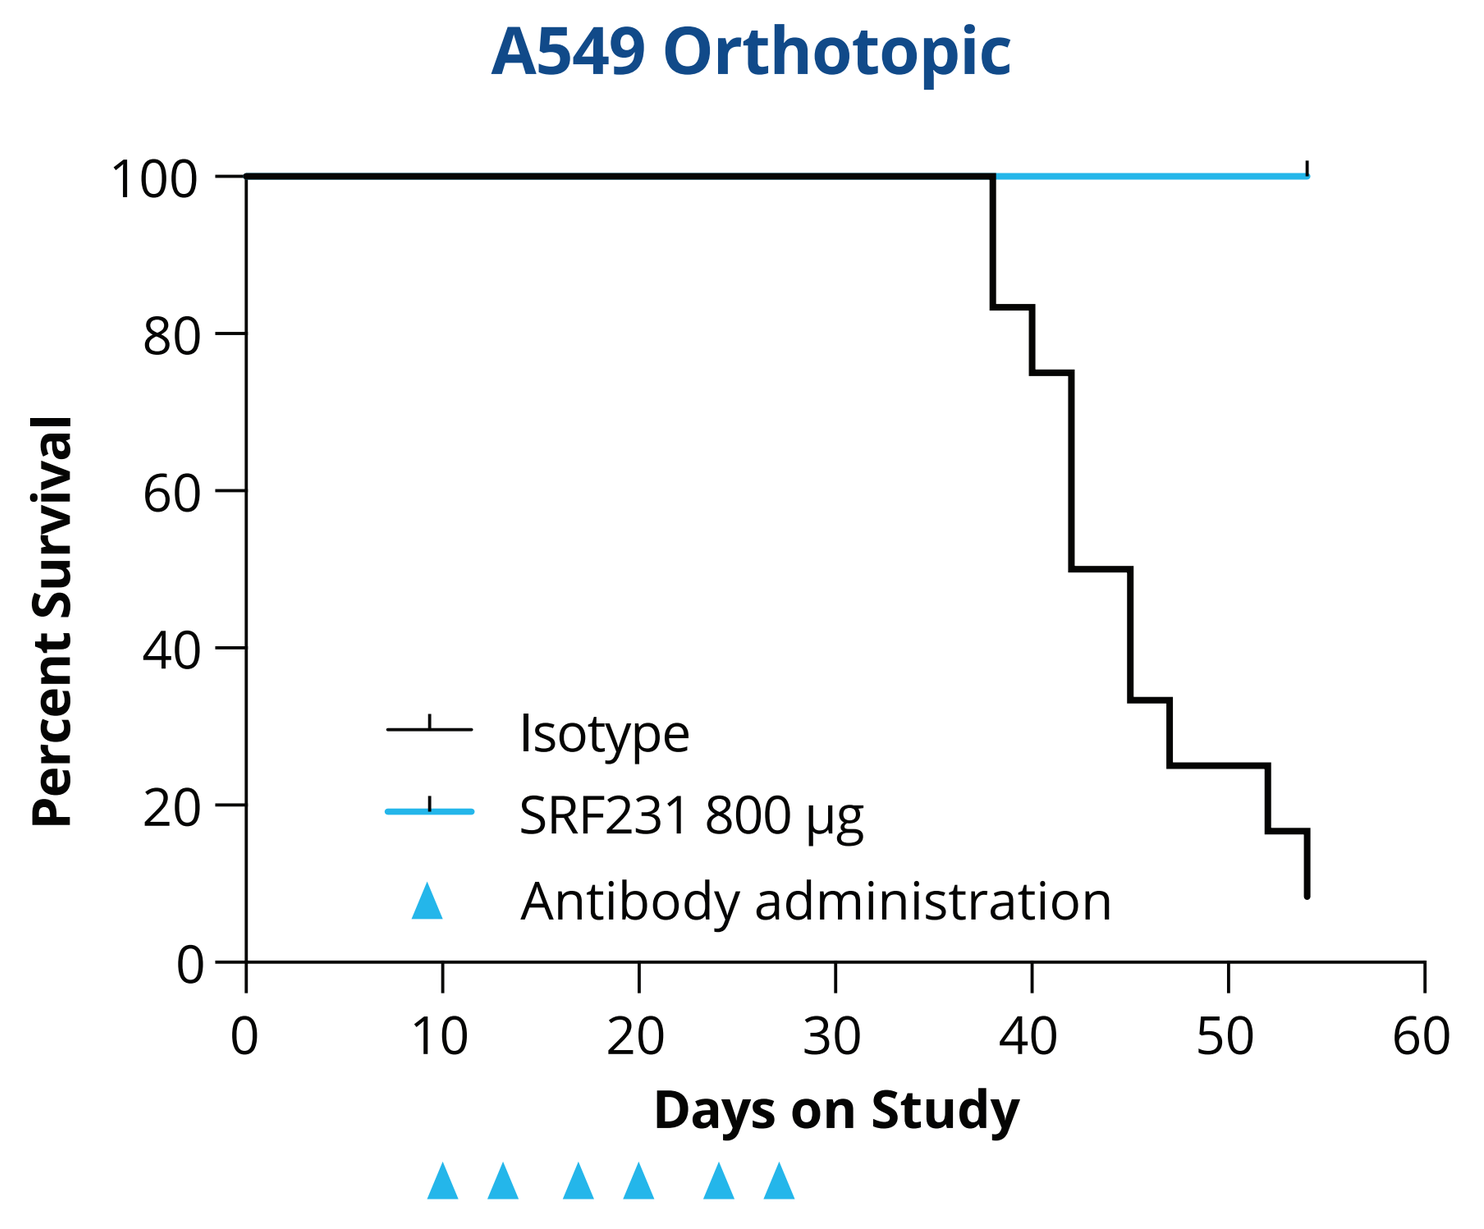
A549 Orthotopic
Percent Survival
Days on Study
Isotype
SRF231 800 µg
Antibody administration

Figure 6: SRF231 Administration Enhances Survival in the A549 Lung Orthotopic Xenograft Model

A549-Luc tumor cells were implanted orthotopically into the lungs of SCID beige mice. Ten days postimplant, animals were sorted based on BLI imaging and treated IP with 800 µg SRF231 or human IgG4 isotype control antibodies biw x 3.

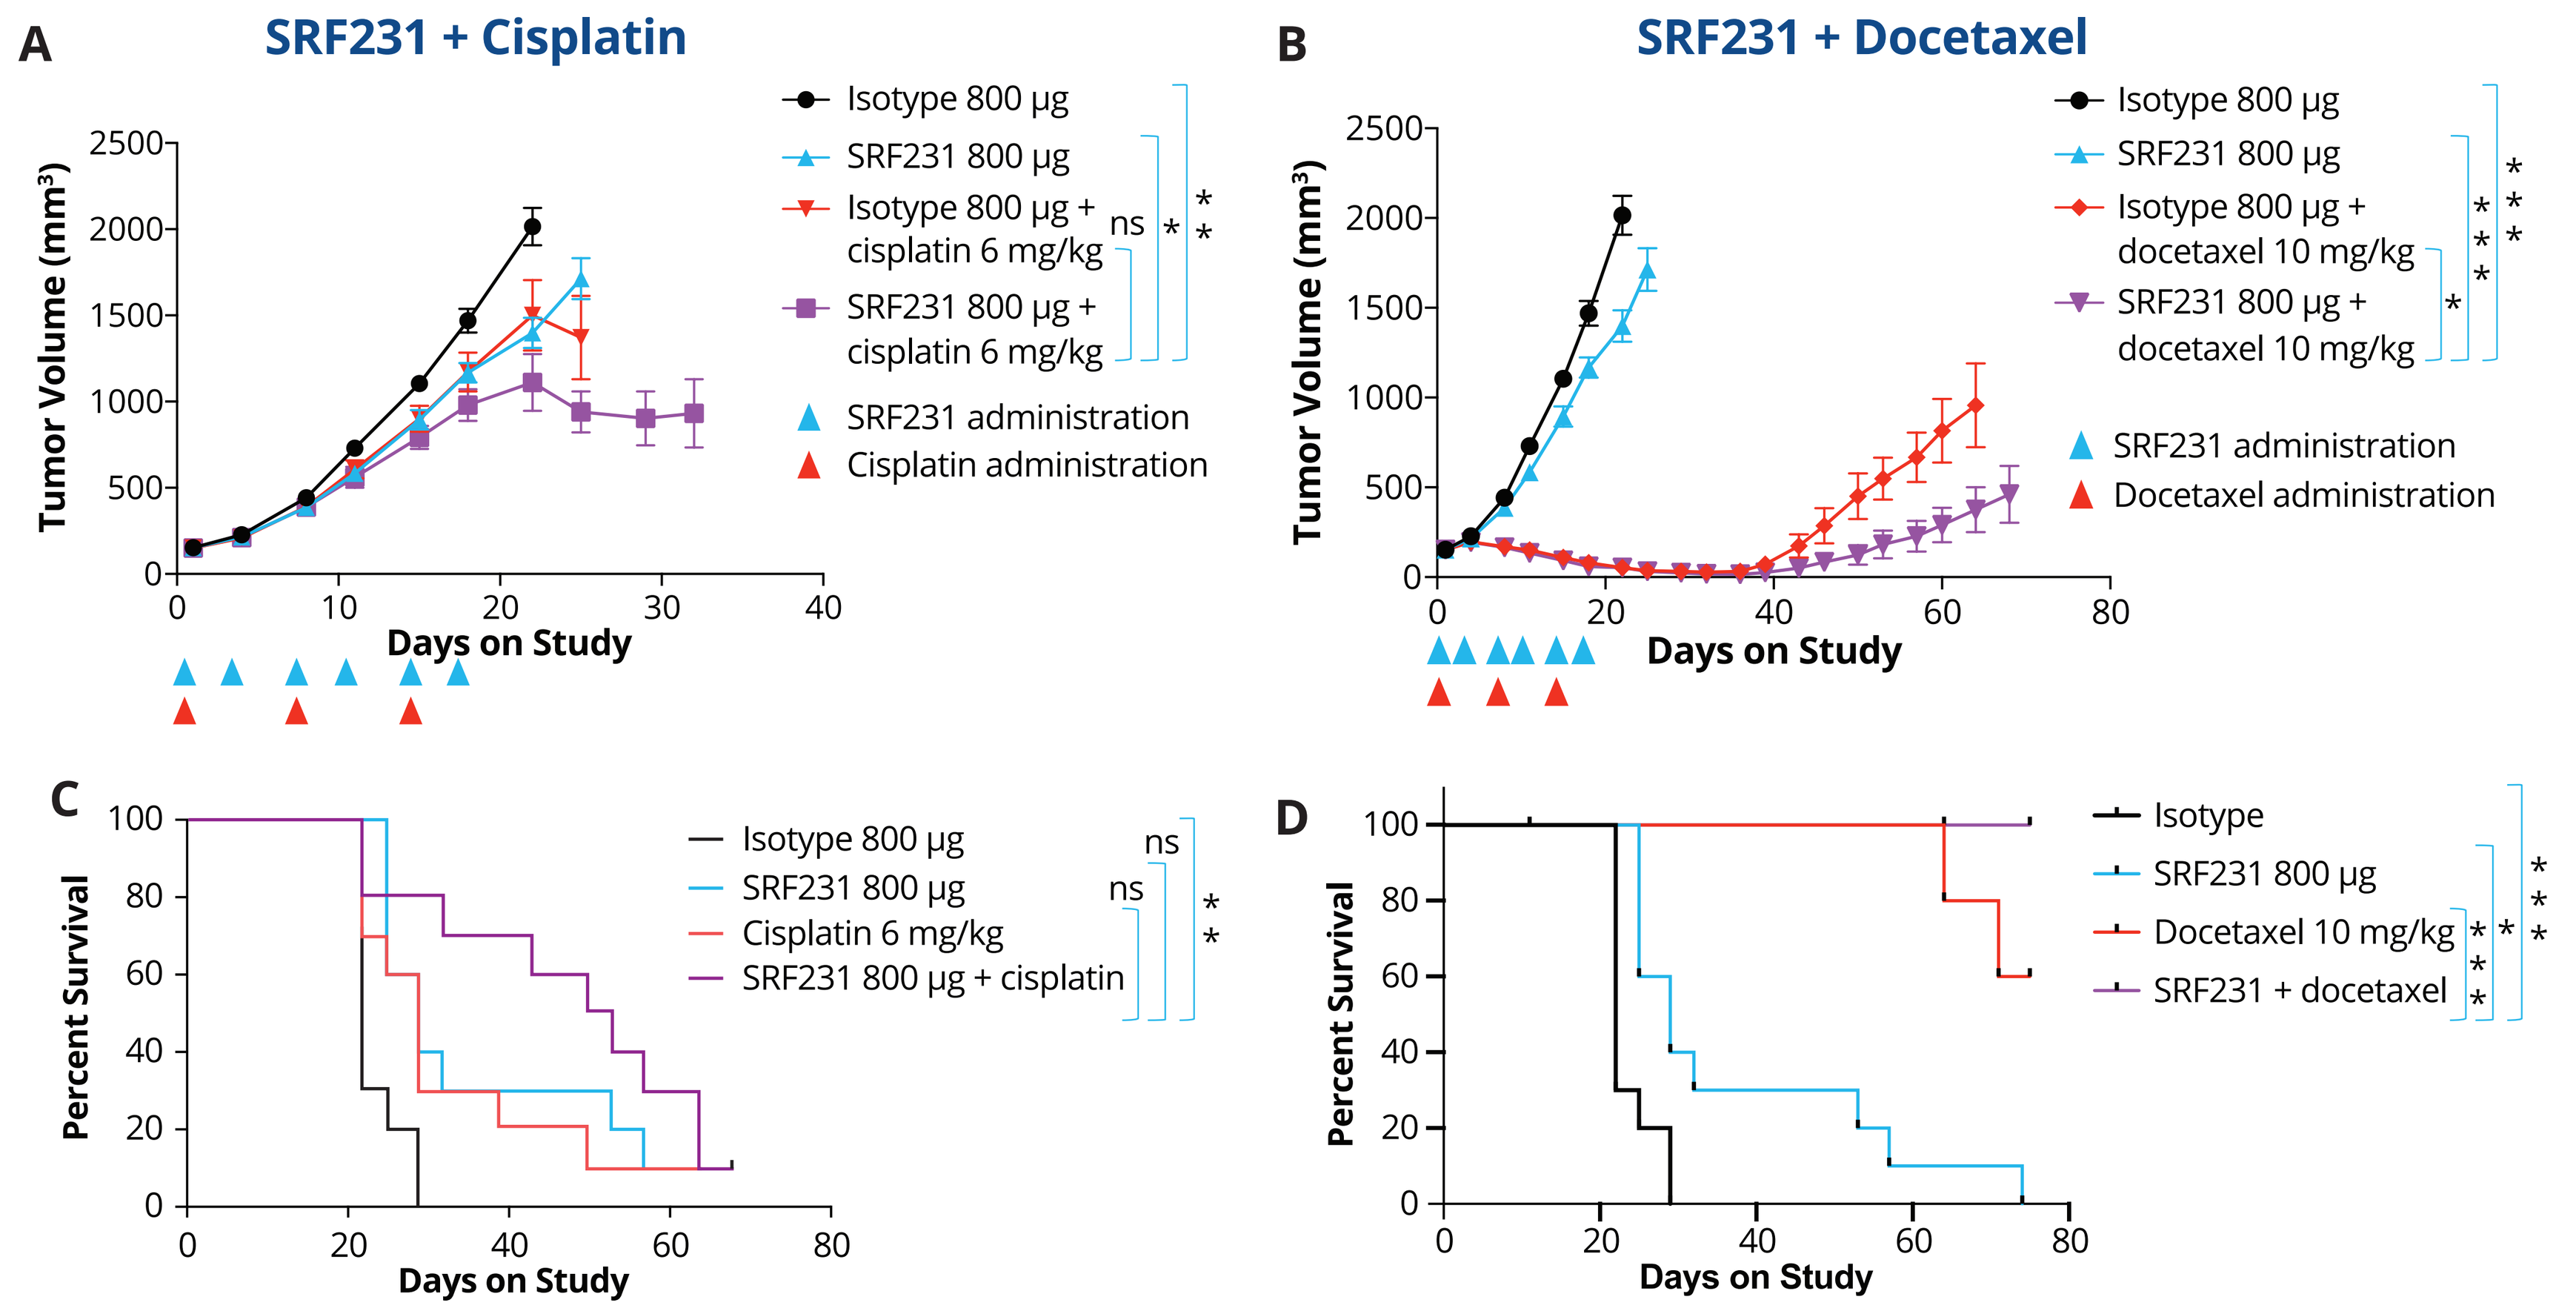
A
SRF231 + Cisplatin
Isotype 800 µg
SRF231 800 µg
Isotype 800 µg + cisplatin 6 mg/kg
SRF231 800 µg + cisplatin 6 mg/kg
SRF231 administration
Cisplatin administration
B
SRF231 + Docetaxel
Isotype 800 µg
SRF231 800 µg
Isotype 800 µg + docetaxel 10 mg/kg
SRF231 800 µg + docetaxel 10 mg/kg
SRF231 administration
Docetaxel administration
C
Isotype 800 µg
SRF231 800 µg
Cisplatin 6 mg/kg
SRF231 800 µg + cisplatin
D
Isotype
SRF231 800 µg
Docetaxel 10 mg/kg
SRF231 + docetaxel
Tumor Volume (mm³)
Percent Survival
Days on Study

Figure 7: Chemotherapy Combined with SRF231 Leads to Enhanced Antitumor Activity in H-1975 (EGFR Mutant) Lung Xenograft Model

Nude mice were implanted SC with NCI-H-1975 cells. Established tumors were then treated IP with 800 µg SRF231 biw x 3. **(A)** + 6 mg/kg cisplatin or **(B)** + 10 mg/kg docetaxel. **(A,B)** Chemotherapies were administered IV qw x 3. **(A)** The antitumor benefit observed in the SRF231 + cisplatin combo arm was significantly different than SRF231 alone (Day 22 & Day 25) while the **(B)** docetaxel combo benefit was significantly different from both single-agent arms (Day 64), along with a 44% CRR in the combo group. **(C,D)** Kaplan-Meier survival plots indicate **(C)** a trend toward extended survival in the SRF231 + cisplatin combo arm **(D)** and a significant benefit in the SRF231 + docetaxel combo arm relative to both single-agent arms: ns = not significant, **P*<0.05, ** *P*<0.001, ****P*<0.0001 (**A,B** unpaired t-test; **C,D** Log-rank Mantel-Cox).

Conclusions

- The anti-hCD47 mAb, SRF231, demonstrates both single-agent and combinatorial activity with opsonizing antibodies and cytotoxic chemotherapies in in vitro and in vivo preclinical hematologic and solid tumor cancer models
- As a single agent, SRF231:
 - Was highly potent in the disseminated MM.1S multiple myeloma xenograft model, and a dose-dependent survival benefit was seen with dosing regimens as low as 3 µg/mouse, qw x 3
 - Extended survival in the orthotopic A549 lung xenograft model
- In combination with opsonizing antibodies or cell death-inducing chemotherapies, SRF231:
 - Potentiated the activity of dara (anti-CD38) and elo (anti-SLAMF7) in multiple myeloma xenograft models, with either single- or multi-dose administration of SRF231, leading to enhanced CRRs and overall survival
 - Enhanced antitumor activity trending towards extended survival in combination with cisplatin and led to delayed tumor regrowth and marked survival benefit in the H-1975 lung xenograft model in combination with docetaxel
 - Please refer to poster #4515 for more on SRF231 combinations

Presented at AACR 2020